Givinostat effects on DMD pathogenesis

Simonetta Andrea Licandro[1], Silvia Consalvi[2], Stefania Petrini[3], Krista Vandenborne[4], Sara Cazzaniga[1], Gianluca Fossati[1], Paolo Bettica[1], Pier Lorenzo Puri[5], Annemieke Aartsma-Rus[6].

[1]Italfarmaco S.p.A., Milan, Italy; [2]UniCamillus International Medical University in Rome, Rome, Italy; [3]Confocal Microscopy Core Facility, Ospedale Pediatrico Bambino Gesù, Rome, Italy; [4]Department of Physical Therapy, University of Florida, Gainesville, Florida, USA; [5]Department of Human Development, Aging and Regeneration Program, Sanford Burnham Prebys Medical Discovery Institute, La Jolla, CA, USA; [6]Department of Human Genetics, Leiden University Medical Center, Leiden, The Netherlands.

INTRODUCTION

Duchenne muscular dystrophy (DMD) is a lethal X-linked disorder that leads to muscle wasting.

The lack of dystrophin results in upregulated HDAC enzymes.

Givinostat, a pan-histone deacetylase inhibitor, significantly reduces fibrosis and promotes the increase of cross sectional area (CSA) in *mdx* skeletal muscles.

Givinostat phase 3 study (Epidys Study) in ambulant DMD boys met its primary endpoint with consistent results in the key secondary endpoints.

OBJECTIVES

The aim of this poster is to show the effect of Givinostat on the DMD pathogenesis and describe its mechanism of action (MoA).

METHODS / DESIGN

***In vivo* Study**

Animals: *mdx* and WT (healthy) C57BL10/J male mice (7 weeks old) were used. Animal experimentation was in compliance with D.lgs 26/2014.

Approach: Givinostat was dissolved in methylcellulose 0.5% and administered p.o. to *mdx* mice (qdx5x15weeks). The efficacy of Givinostat was evaluated by grip test (www.treat-nmd.eu/research/preclinical/dmd-sops/). Microscopic examinations of tibialis anterior (TA), gastrocnemius (GAS) and diaphragm (DIA) muscles were performed on formalin-fixed paraffin-embedded sections stained by H&E and Sirius Red or O Red Oil staining (only for TA).

Clinical Trials

Study 43: open label 2-part, phase 2 clinical trial, which enrolled 20 ambulant DMD boys aged 7 to <11 years. Boys were on a stable dose of corticosteroids for ≥ 6 months[3]. The primary endpoint was the change in histology parameters comparing the brachial biceps biopsies before and after at least 12 months of treatment with givinostat (NCT01761292).

Epidys Study: Phase 3 Randomized, double blind, parallel group, placebo-controlled study in ambulant DMD boys, on stable corticosteroids for ≥ 6 months, to demonstrate that givinostat preserves muscle mass and slows down disease progression (NCT02851797).

KEY POINTS

- Givinostat increase Cross sectional area (CSA), and reduced fibrosis, necrosis and fat replacement in skeletal muscle tissue in both *mdx* mice and DMD boys.
- Givinostat significantly reduces the decline of muscle function in *mdx* mice and in DMD ambulant boys.

CONCLUSIONS

- Givinostat was shown to counteract all the pathogenetic events downstream of the lack of dystrophin in DMD mouse model and in DMD boys.
- Combined, these effects result in reduced muscle deterioration and function decline.

RESULTS

Histology Results in *mdx* mice

- Givinostat at 37.5 mg/kg dose significantly increased the CSA in GAS muscle (Figure 1)[1].
- Givinostat significantly reduced fat deposition starting from 5 mg/kg dose in TA muscle (**p<0.01 vs Vehicle; 1-way ANOVA with Bonferroni's multiple comparison test (Figure 2)[2].
- Givinostat significantly prevented fibrosis starting from 1 mg/kg dose in TA, GAS and DIA muscles, respectively (*p<0.05; **p<0.01; ***p<0.001; ****p<0.0001 vs Vehicle; 1-way ANOVA with Bonferroni's multiple comparison test (Figure 3)[1].

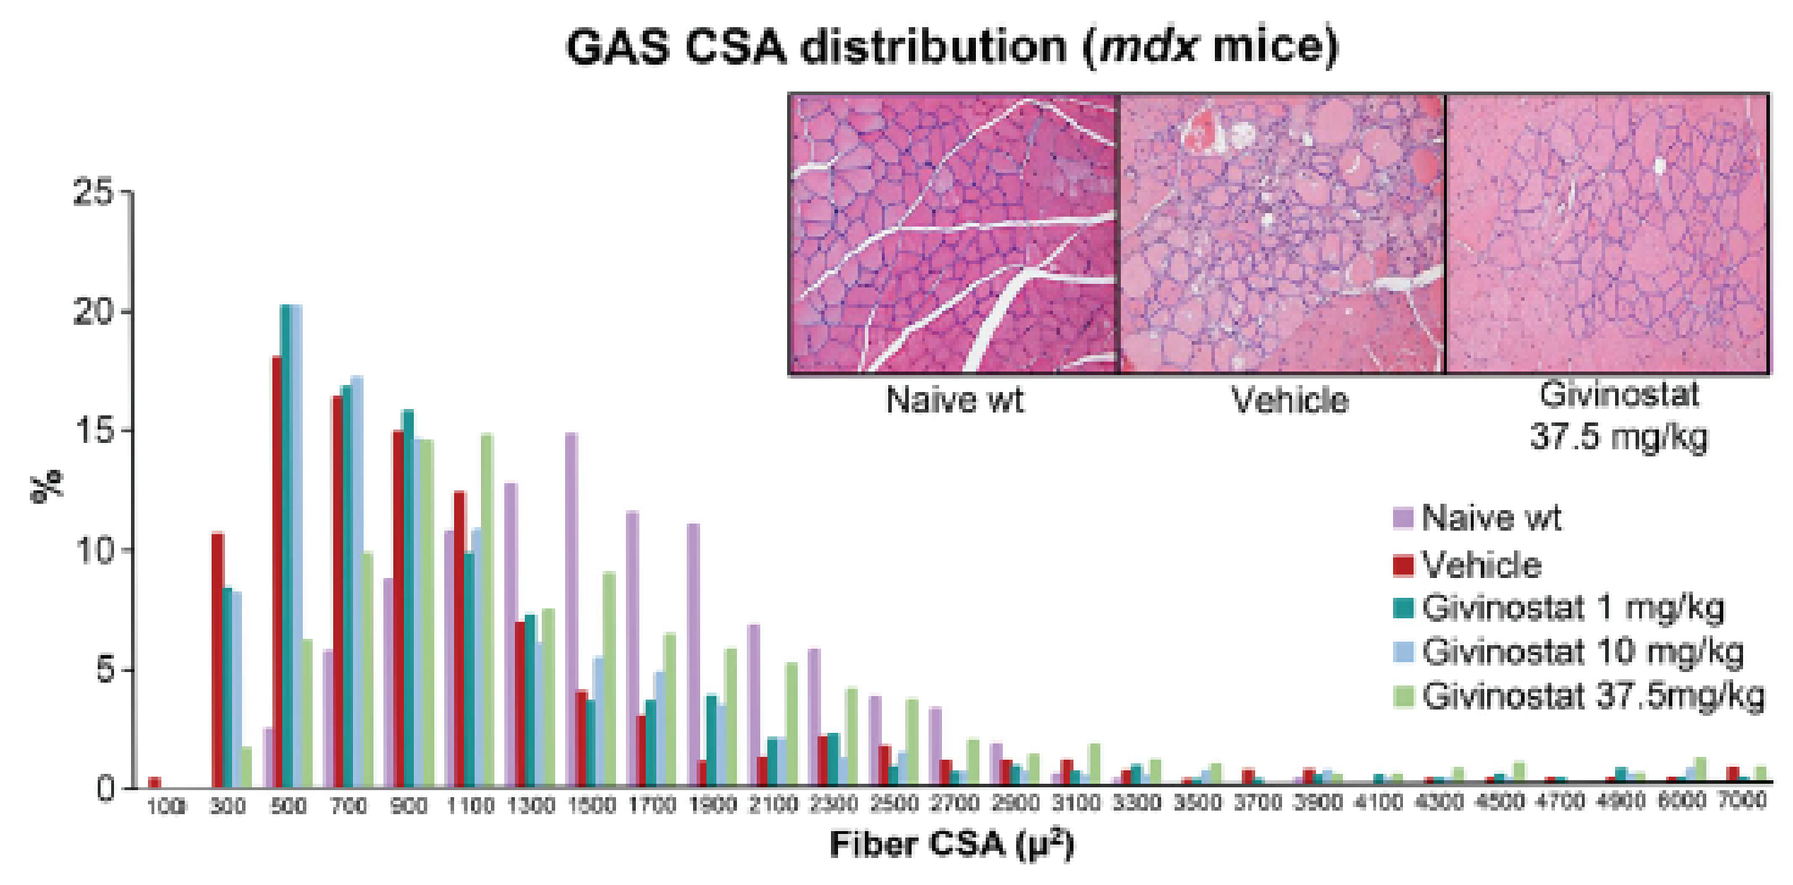
GAS CSA distribution (mdx mice)
Naive wt
Vehicle
Givinostat 37.5 mg/kg
Naive wt
Vehicle
Givinostat 1 mg/kg
Givinostat 10 mg/kg
Givinostat 37.5mg/kg
%
Fiber CSA (μ²)

Figure 1 – Fiber CSA different size distributions in GAS muscle (N=100 myofibers; 5 mice/group).

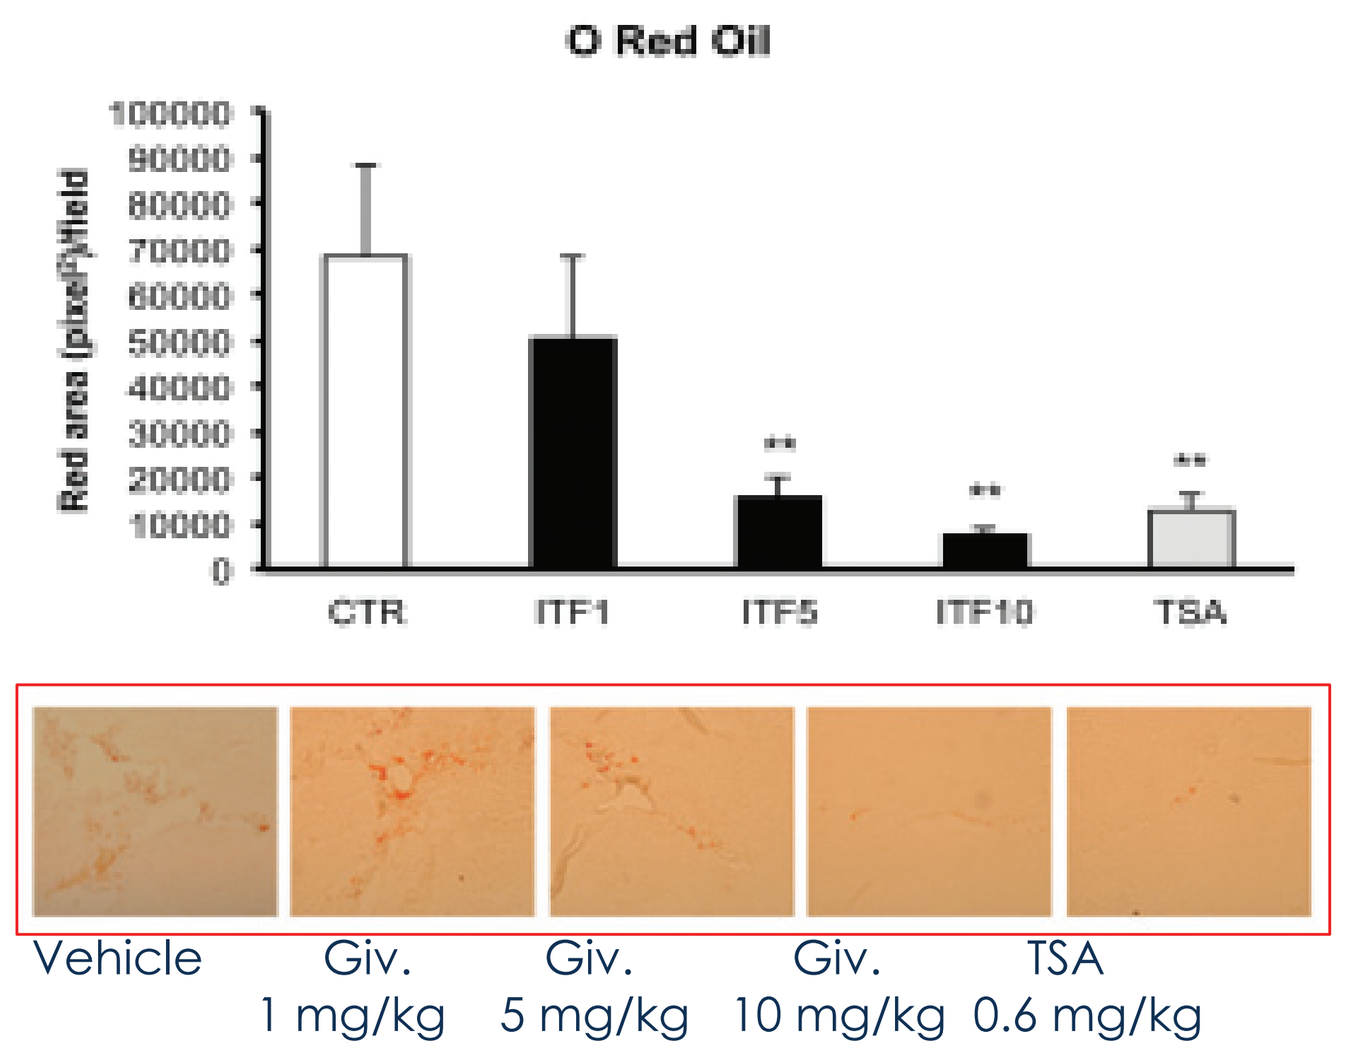
O Red Oil
Red area (pixel²)/field
CTR
ITF1
ITF5
ITF10
TSA
Vehicle
Giv. 1 mg/kg
Giv. 5 mg/kg
Giv. 10 mg/kg
TSA 0.6 mg/kg

Figure 2 – Fat accumulation in *mdx* muscles (N=5; Giv.=Givinostat).

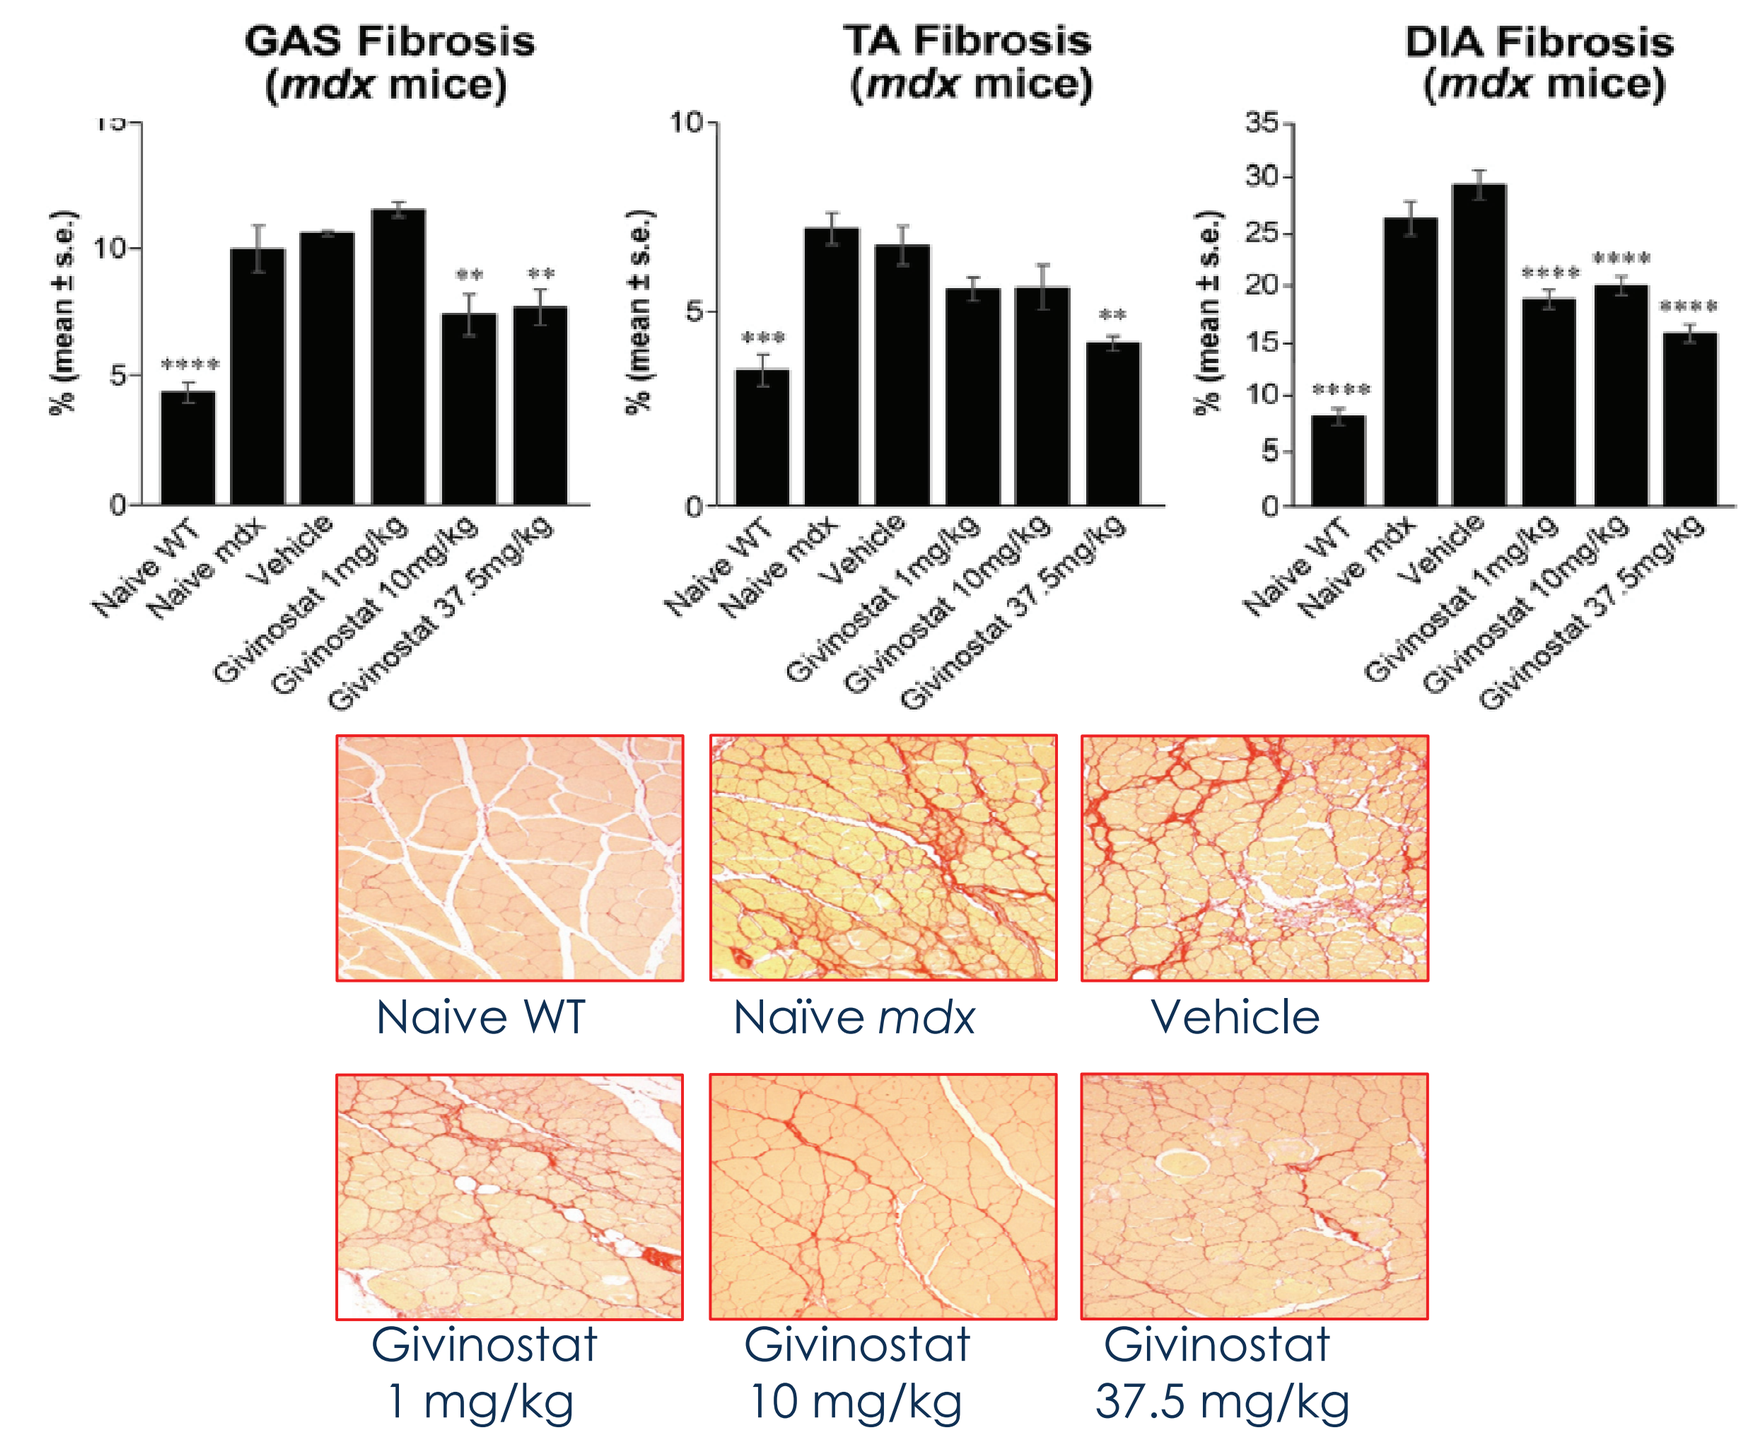
GAS Fibrosis (mdx mice)
TA Fibrosis (mdx mice)
DIA Fibrosis (mdx mice)
% (mean ± s.e.)
Naive WT
Naive mdx
Vehicle
Givinostat 1mg/kg
Givinostat 10mg/kg
Givinostat 37.5mg/kg
Naive WT
Naïve mdx
Vehicle
Givinostat 1 mg/kg
Givinostat 10 mg/kg
Givinostat 37.5 mg/kg

Figure 3 – Fibrosis percentage in *mdx* muscles (N=5).

Functional Results in *mdx* mice

- Givinostat 10 mg/kg represented the minimum effective dose that showed a statistically significant increase in maximum normalized strenght (FNmax) values until the end of treatments in *mdx* mice (*p<0.05; **p<0.01; ***p<0.001; ****p<0.0001 vs Vehicle at different time points) (Figure 4)[1].
- Mice treated with 25 and 37.5 mg/kg doses of Givinostat reached FNmax values comparable to those of healthy mice (from day 49 to day 65)[1].

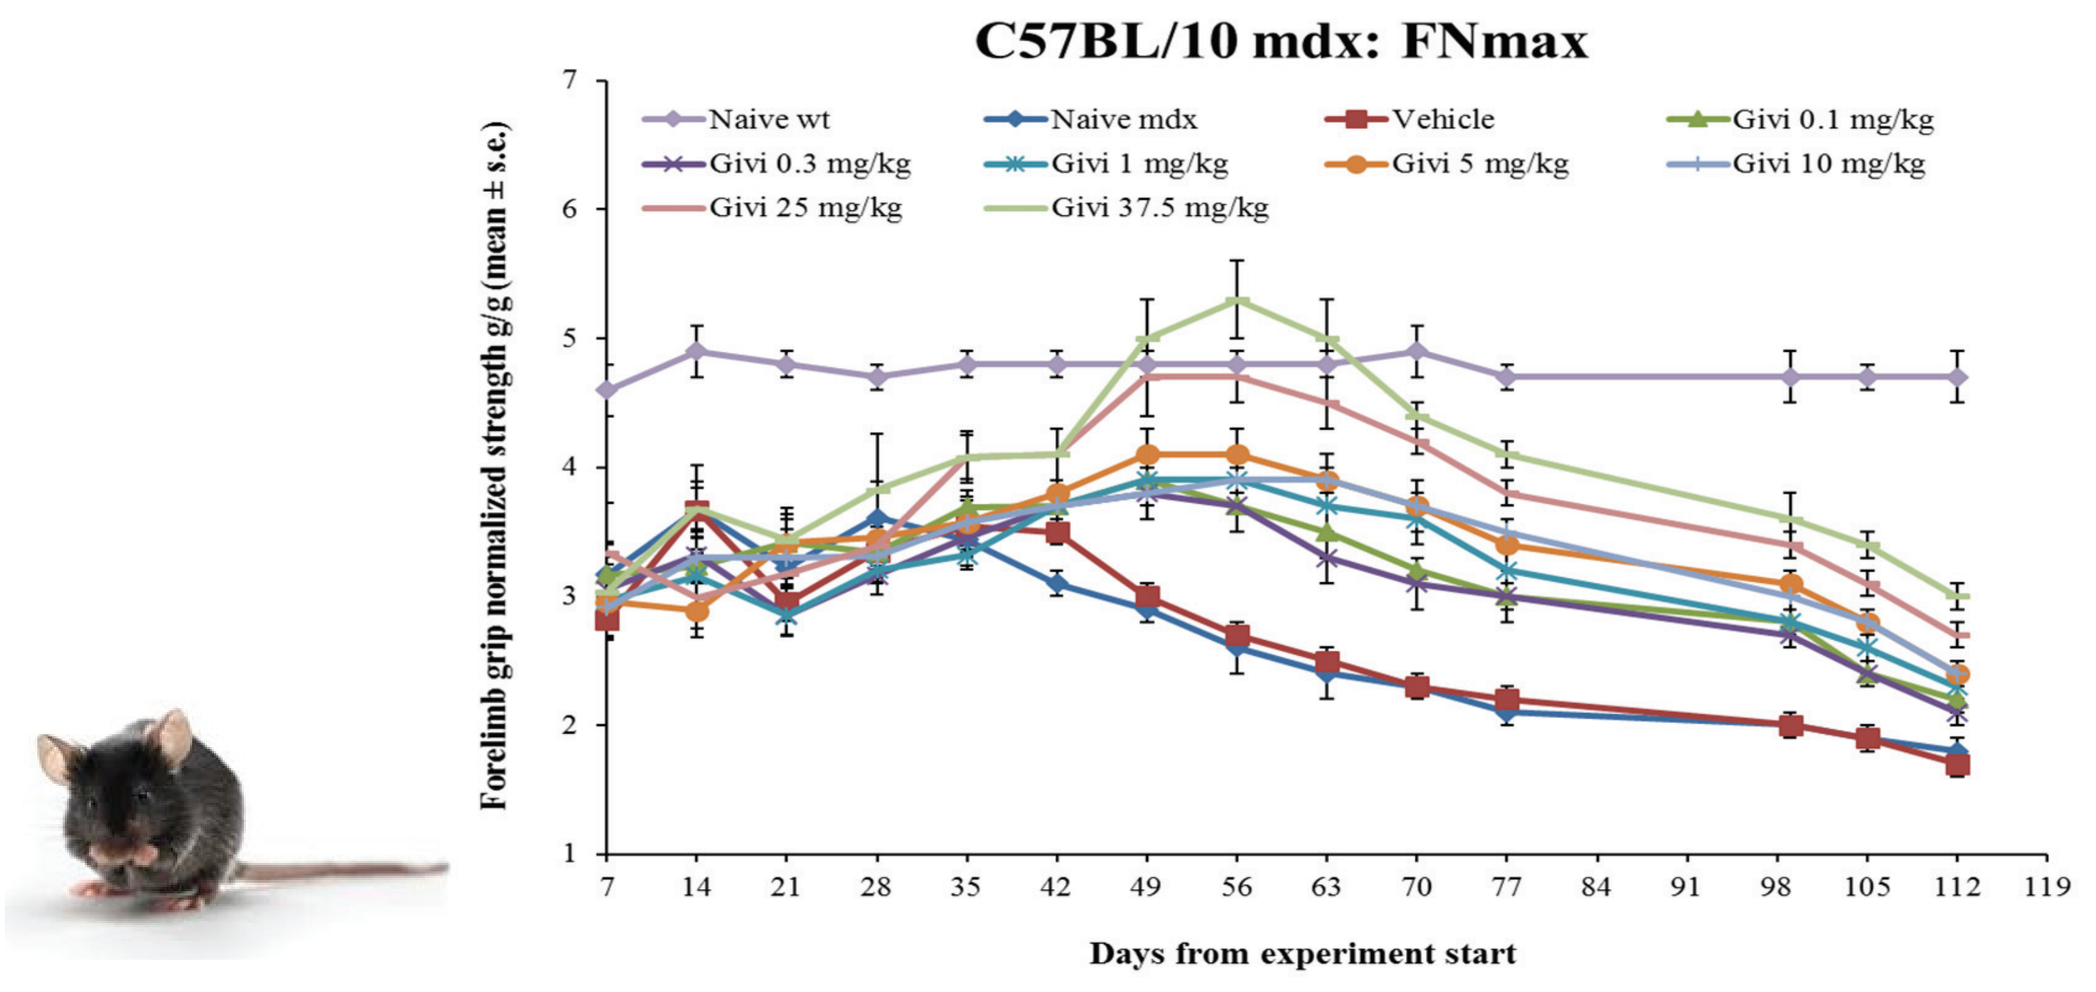
C57BL/10 mdx: FNmax
Naive wt
Naive mdx
Vehicle
Givi 0.1 mg/kg
Givi 0.3 mg/kg
Givi 1 mg/kg
Givi 5 mg/kg
Givi 10 mg/kg
Givi 25 mg/kg
Givi 37.5 mg/kg
Forelimb grip normalized strength g/g (mean ± s.e.)
Days from experiment start

Figure 4 – Grip test results (N=9).

Study 43

- **Histology Results in DMD boys** – the amount of muscle (MFAF) and the Cross Sectional Area (CSA) of the muscle fibers were significantly increased at the end of the study, while the amount of fibrotic tissue (Total Fibrosis), Necrosis and Fatty Replacement significantly decreased (Figure 5)[3].

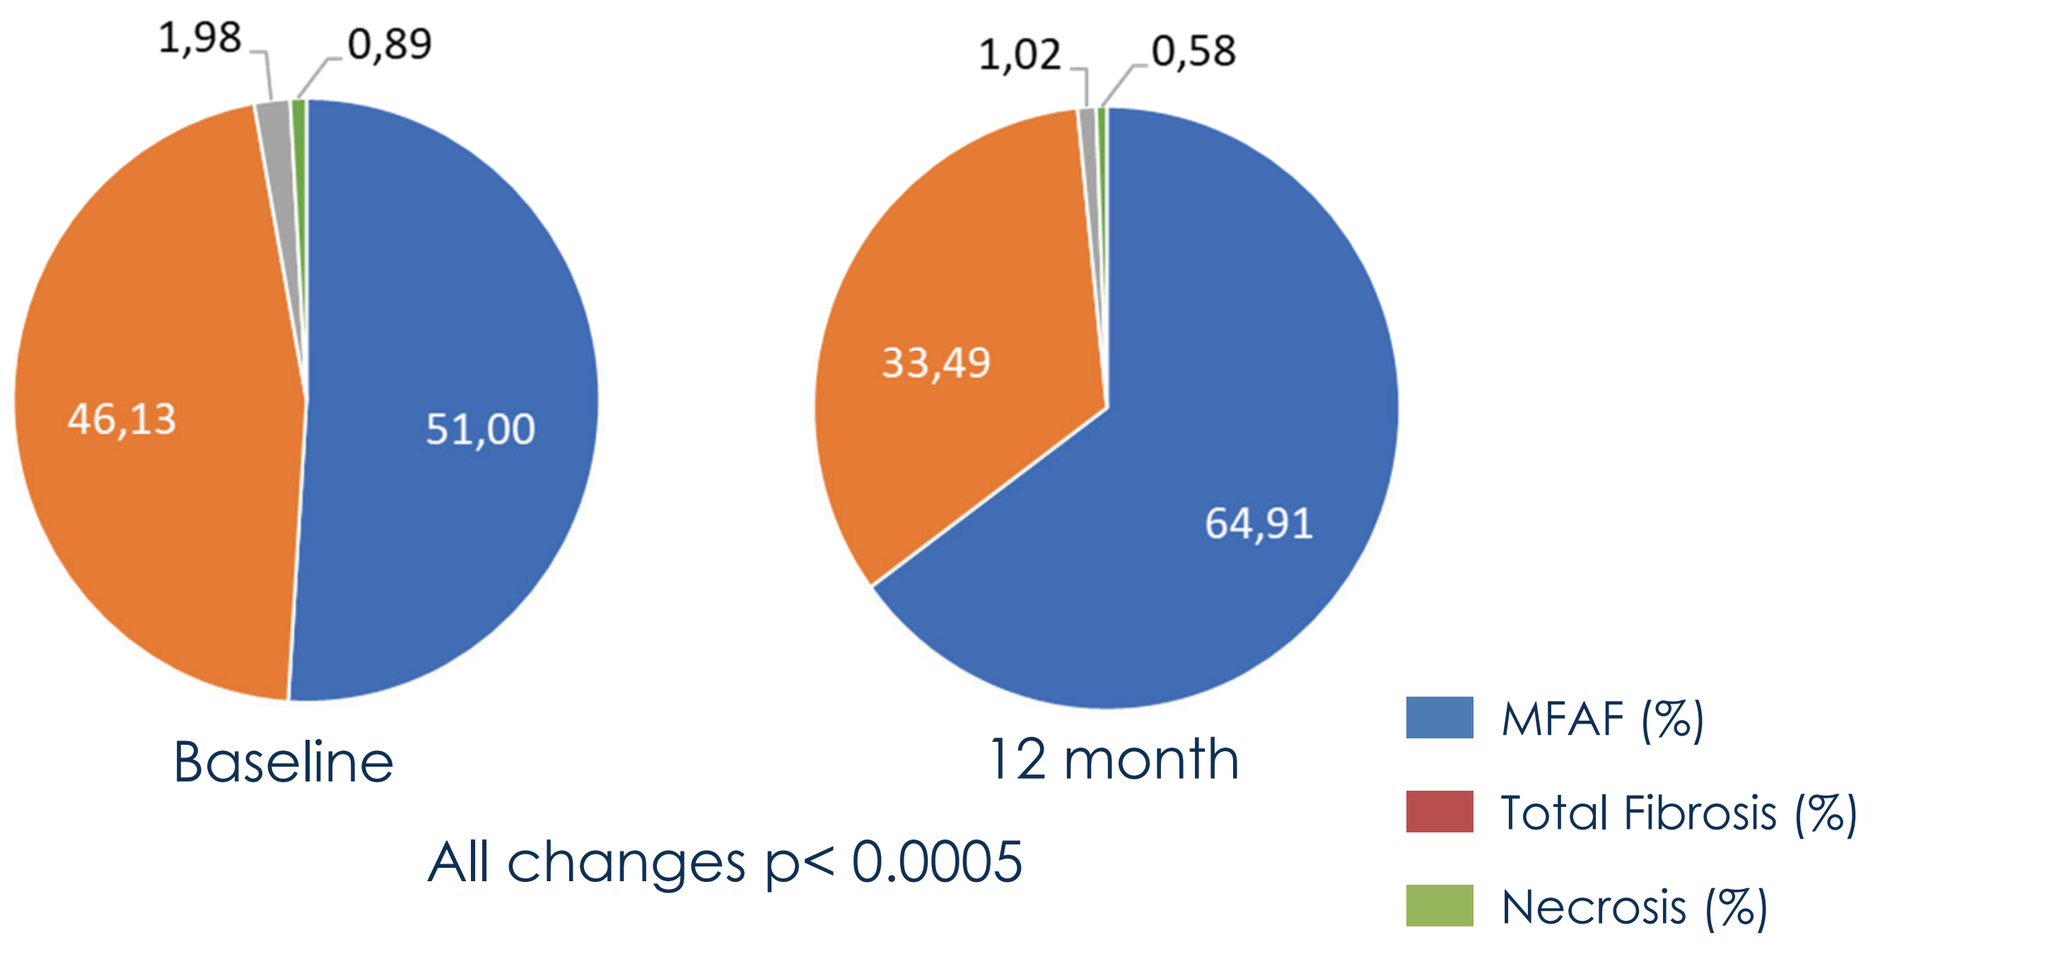
1,98
0,89
46,13
51,00
1,02
0,58
33,49
64,91
Baseline
12 month
All changes p< 0.0005
MFAF (%)
Total Fibrosis (%)
Necrosis (%)
Fatty Replacement (%)

Figure 5 – Histology results (N=18).

Study 48

- **Magnetic Resonance Results in DMD boys** – Givinostat reduced by ≈30% fatty infiltration of vastus lateralis muscle key for ambulation (Figure 6). Magnetic Resonance Spectroscopy – MRS – and Dixon MRI results were similar[4].
- **Serum Biomarker in DMD boys** – Givinostat reduced circulating biomarkers of fibrosis (TGFbeta) and inflammation (IP10) (Figure 7)[5].

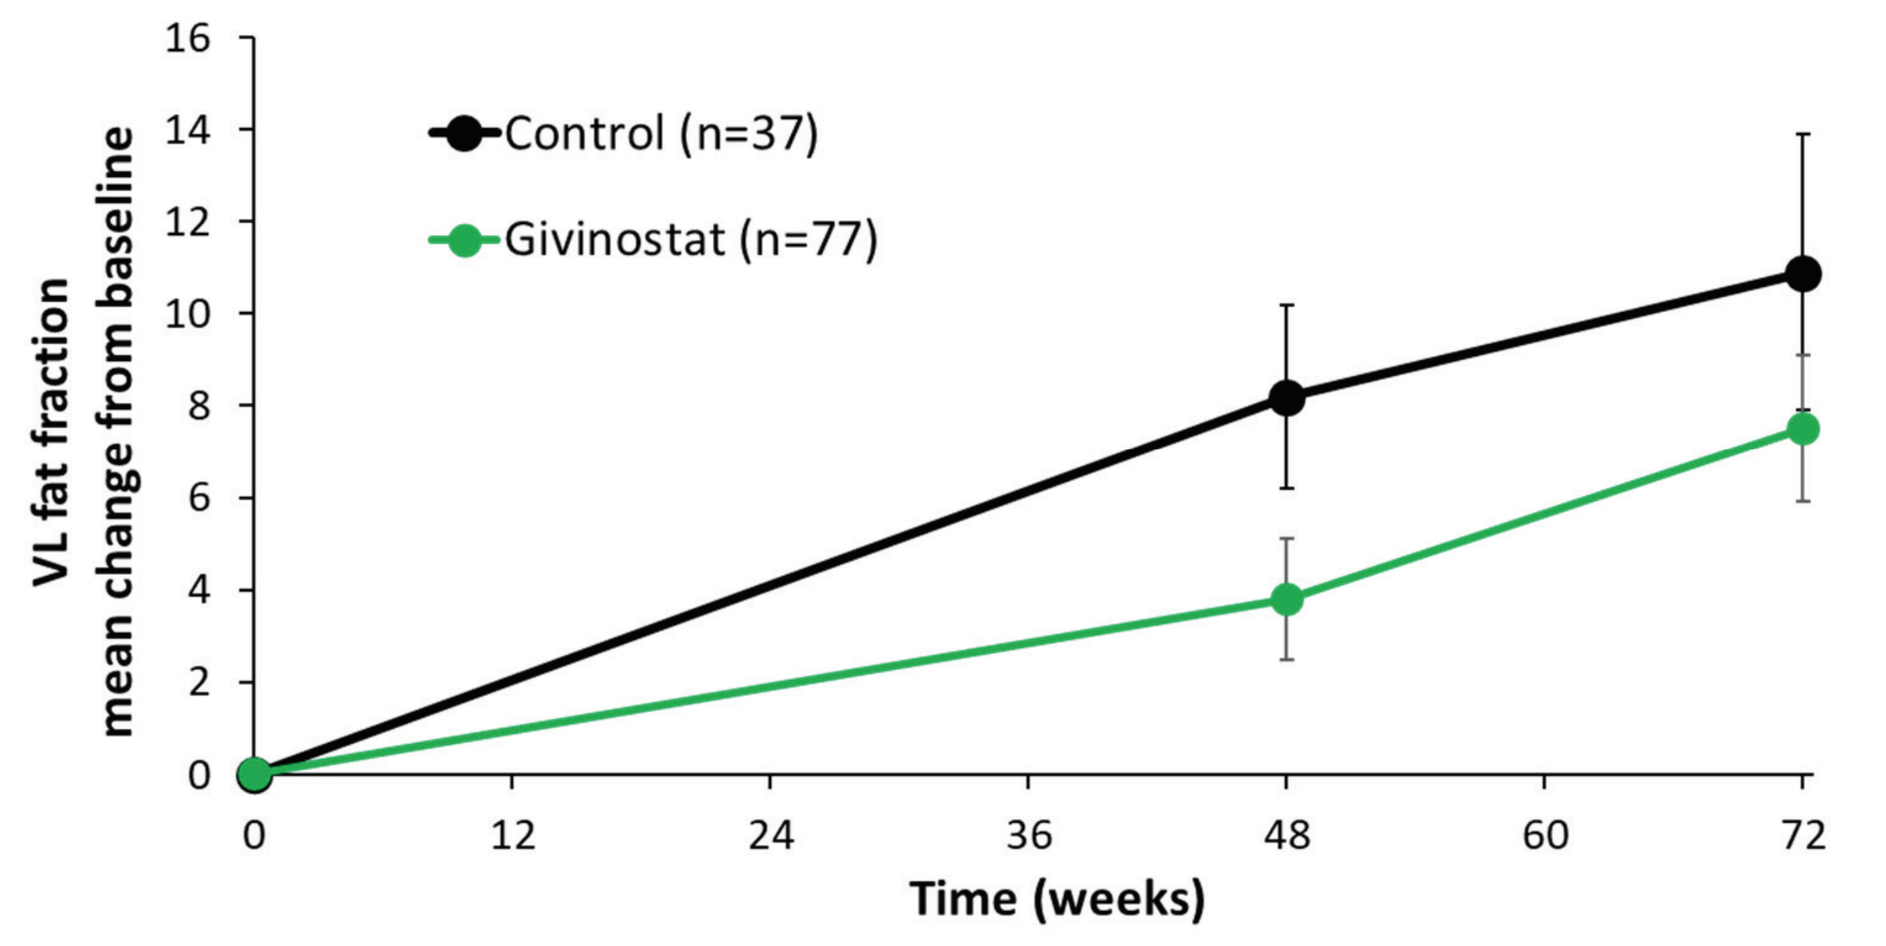
Control (n=37)
Givinostat (n=77)
VL fat fraction mean change from baseline
Time (weeks)

Figure 6 – Mean Change of Vastus Lateralis Fat fraction assessed by MRS.

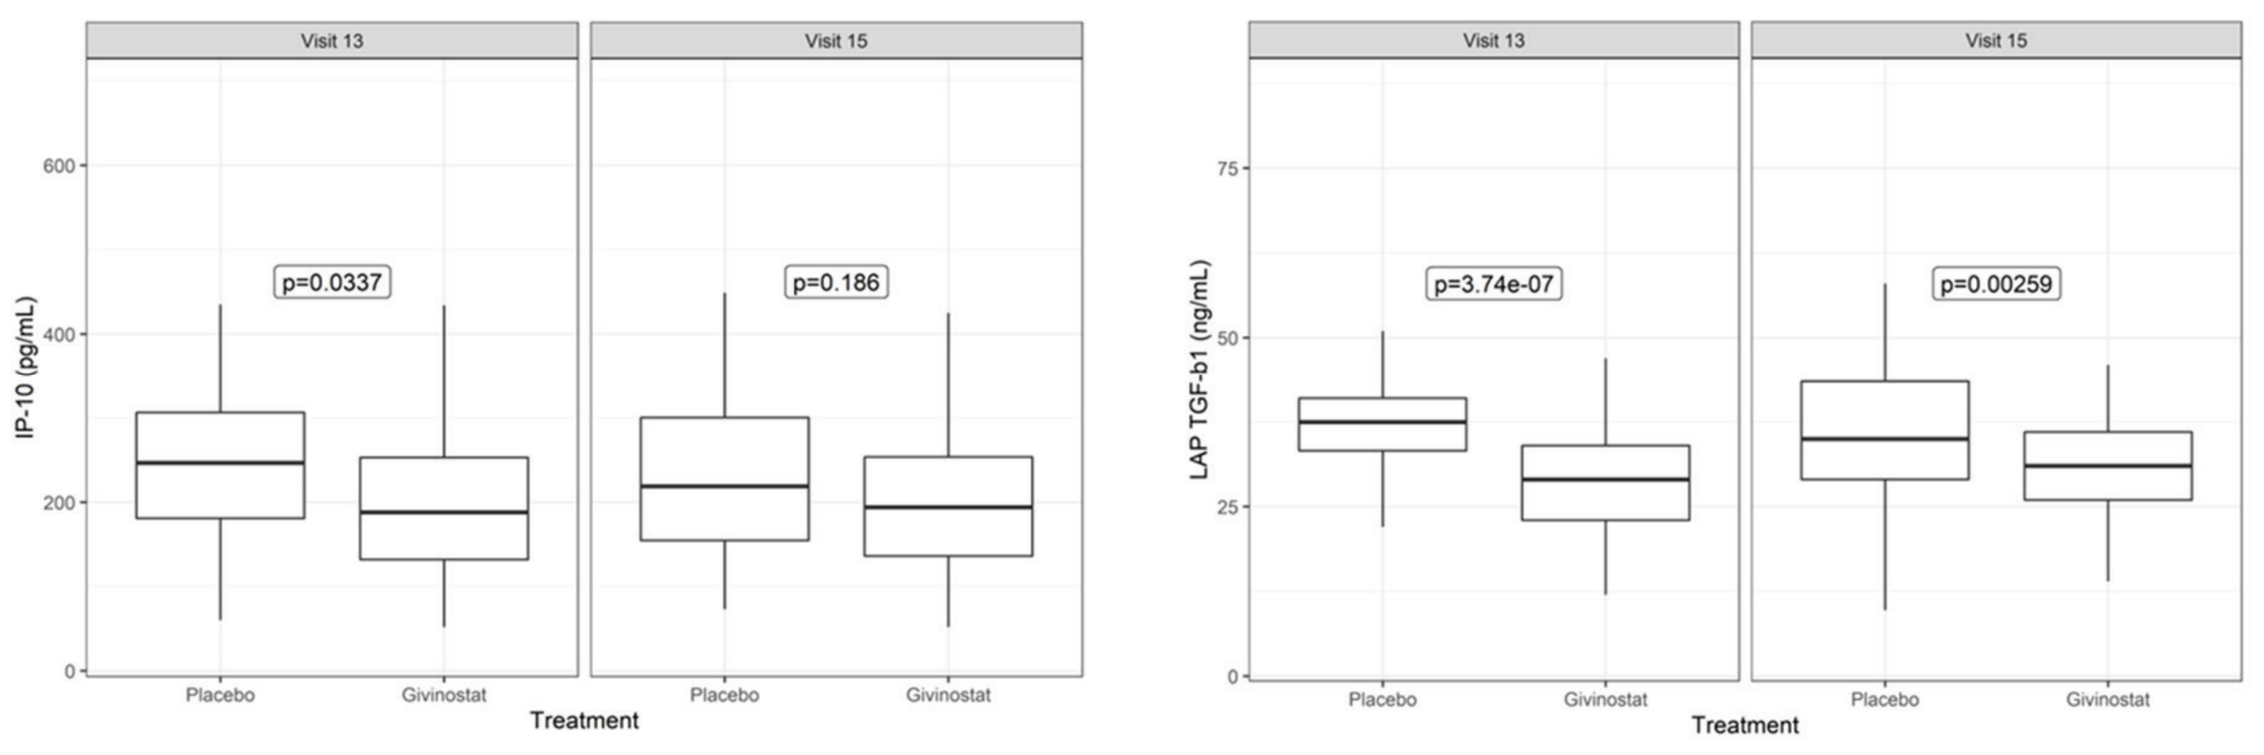
Visit 13
Visit 15
p=0.0337
p=0.186
p=3.74e-07
p=0.00259
IP-10 (pg/mL)
LAP TGF-b1 (ng/mL)
Placebo
Givinostat
Treatment

Figure 7 - Serum Biomarker (Givinostat N=114; placebo N=58).

Functional Results in DMD boys

- In Phase 3 Epidys trial, Givinostat significantly reduces the decline (i.e. increase) in time to perform 4 Stairs Climb - 4SC (primary endpoint) by ~40%.
- Over time, Givinostat consistently reduces decline in muscle function and strength (secondary key endpoints) (Figure 8)[4].

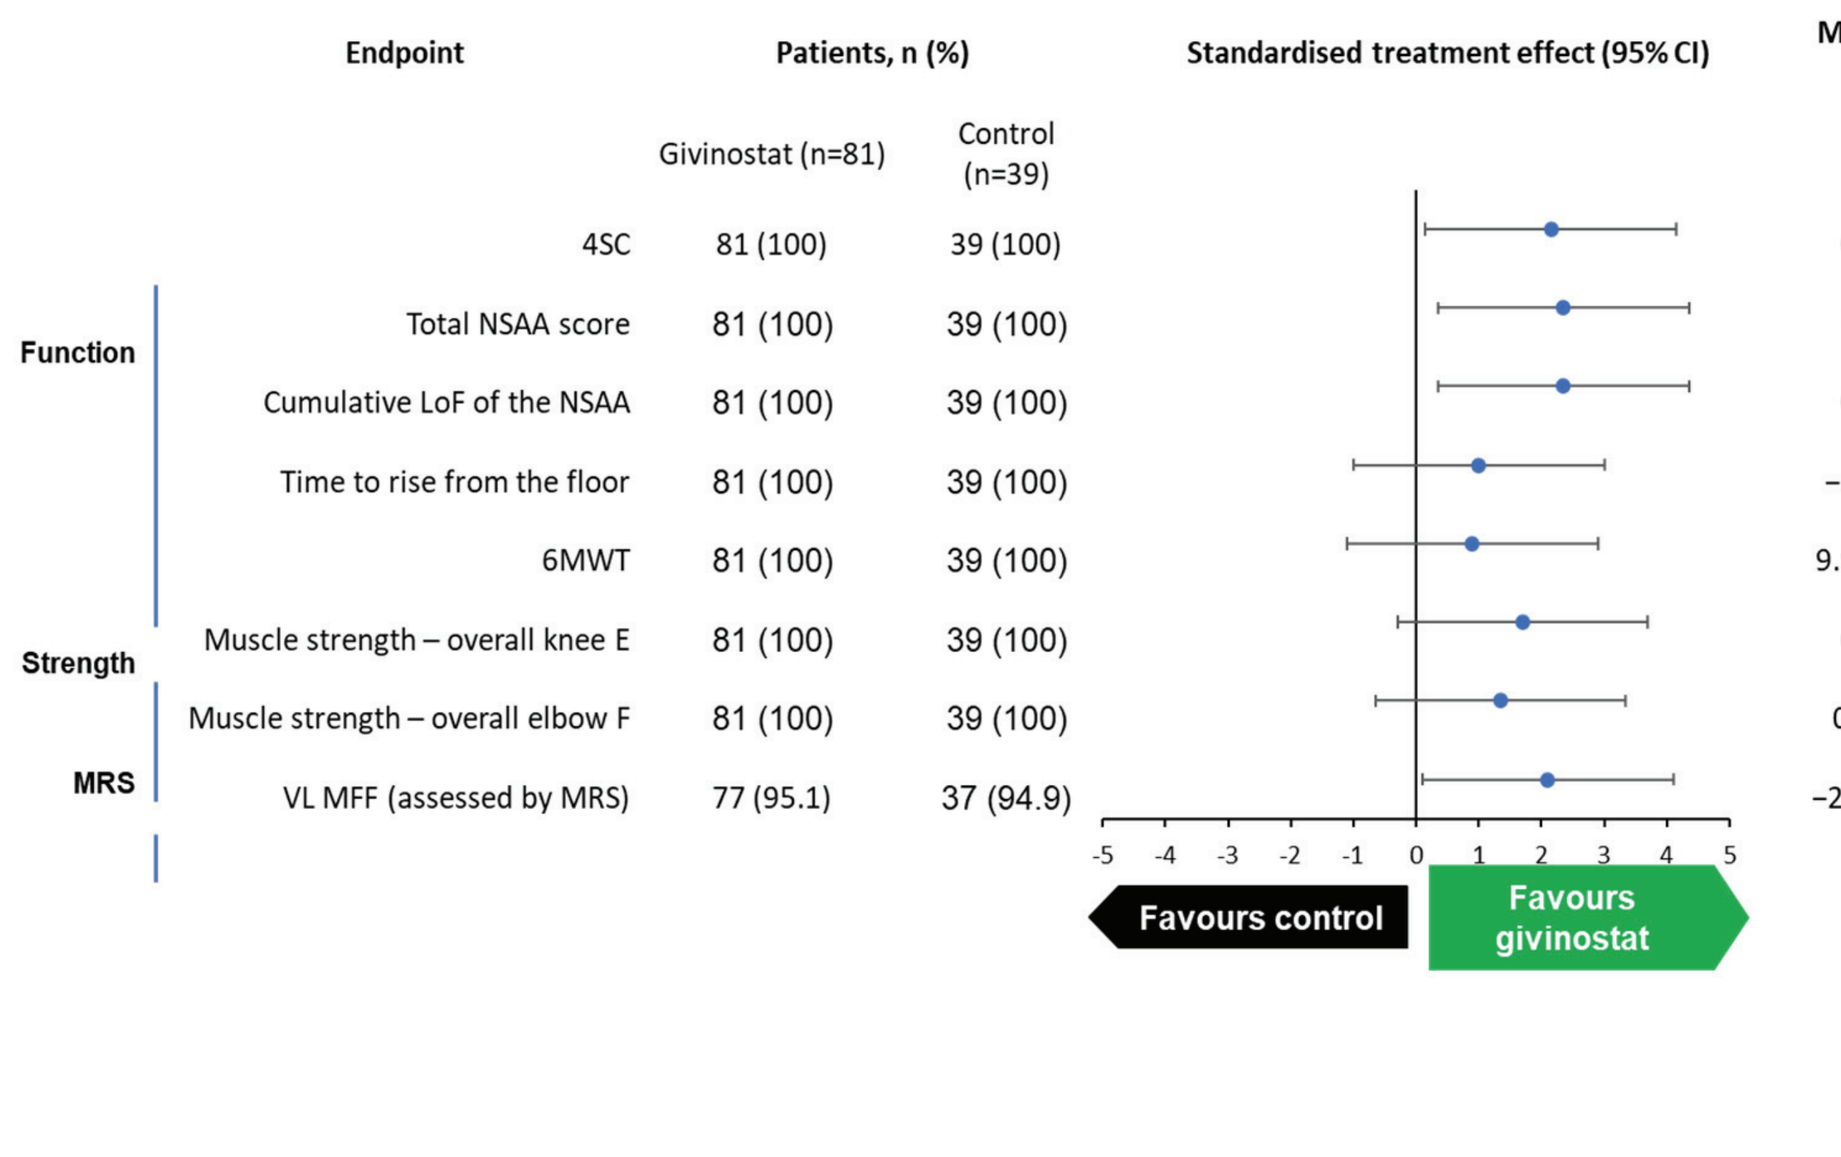
Standardised treatment effect (95% CI)
Favours control
Favours givinostat

	Endpoint	Patients, n (%) Givinostat (n=81)	Control (n=39)	Mean difference (95% CI)	p-value
Function	4SC	81 (100)	39 (100)	0.86 (0.745, 0.989)	0.0345
	Total NSAA score	81 (100)	39 (100)	1.91 (0.295, 3.533)	0.0209
	Cumulative LoF of the NSAA	81 (100)	39 (100)	0.61 (0.408, 0.927)	0.0202
	Time to rise from the floor	81 (100)	39 (100)	−3.28 (−9.573, 3.018)	0.03044
	6MWT	81 (100)	39 (100)	9.96 (−12.071, 31.893)	0.3723
Strength	Muscle strength – overall knee E	81 (100)	39 (100)	0.19 (−0.03, 0.401)	0.092
	Muscle strength – overall elbow F	81 (100)	39 (100)	0.09 (−0.041, 0.213)	0.1818
MRS	VL MFF (assessed by MRS)	77 (95.1)	37 (94.9)	−2.92 (−53641, −0.204)	0.0354

Figure 8 - Epidys Trial results.

REFERENCES & ACKNOWLEDGEMENTS

1. Licandro S.A. et al. Skelet Muscle. 2021;11(1):19; 2. Consalvi S. et al. Mol Med. 2013;19(1):79-87; 3. Bettica P. et al. Neur Disord. 2016;26:643-649. 4. Mercuri E. et al. P37, 2023 WMS Conference, Charleston, USA, October 2023. 5. CSR Epidys study.

The authors thank all the study participants and their families, as well as the study team members, at participating Epidys sites for assistance in collecting and managing the data.

Corresponding author contact details:
s.licandro@italfarmacogroup.com

ITALFARMACO

Presented at the 2024 MDA Clinical & Scientific Conference, Orlando, USA, 3-6 March 2024